Efficacy of a new nasal steroid, S0597, for the treatment of ragweed induced allergic rhinitis; tested in an Environmental Exposure Unit (EEU).

Anne K. Ellis, MD MSc FRCPC[1,2], Lisa M. Steacy, BSc[2], Terry J Walker, BA[2], Abhijeet Joshi B Pharm, MBA[3], Shravanti Bhowmik MD[3], and Atul Raut, MD, PhD[3]

1-Department of Medicine, Queen's University, Kingston ON Canada, 2- Allergy Research Unit, Kingston General Hospital, Kingston, ON Canada 3- Sun Pharma Advanced Research Company Ltd., Mumbai, India

Background

Allergic rhinitis (AR) is a significant public health concern worldwide, affecting an estimated 500 million people. **Intranasal glucocorticosteroids (INS)** are regarded as the most effective pharmacologic treatment for AR and are recommended by the Allergic Rhinitis and its Impact on Asthma guidelines as a first-line therapy. INS have a good safety profile with a low risk of systemic side effects due to their low systemic bioavailability. This is especially true for newer INS like ciclesonide, mometasone furoate, and fluticasone furoate, which all have systemic bioavailability of less than 1%.

S0597 is a novel glucocorticosteroid that has been formulated as an intranasal spray for the treatment of seasonal allergic rhinitis (SAR). The short form of its International Union of Pure and Applied Chemistry name is SFDAC. Results of a phase 1, multiple ascending dose study indicated that intranasal S0597 was safe and well tolerated at doses up to 3200 µg once daily. No clinically significant suppression of the hypothalamic–pituitary–adrenal (HPA) axis was observed and the plasma concentration of S0597 was below the lower limit of quantification at most time points for all participants.

The **aim** of the present study was to assess the clinical efficacy, safety, and tolerability of 3 once-daily doses (50, 200, and 400 µg) of S0597 intranasal spray compared with placebo in participants with SAR who were exposed to ragweed pollen challenges in an environmental exposure unit (EEU).

Methods

Study Design: This was a phase 2, randomized, double-blind, placebo-controlled, single-center, 4-arm, parallel study conducted in the EEU at Kingston General Hospital, Kingston, Ontario, Canada, between October 2 and December 17, 2014. Adults with documented SAR during ragweed pollen season were exposed to controlled levels of ragweed pollen in the EEU.

The study was given ethical clearance by the Queen's University Health Sciences and Affiliated Teaching Hospitals Research Ethics Board. The ClinicalTrials.gov identifier is NCT01940146.

Inclusion criteria: Adult participants (18–65 y) were required to have a minimum 2-year history of SAR due to ragweed pollen, a positive skin prick test to short ragweed (defined as a wheal ≥3 mm larger than the negative control) within 12 months prior to enrollment, a total nasal symptom score (TNSS) of at least 6 at least once during a 1- to 3-hour screening/priming challenge in the EEU and within the first 2 hours of the baseline visit, and forced expiratory volume in 1 minute (FEV1) of at least 80% of predicted during screening. **Exclusion criteria:** symptomatic perennial AR; clinically significant nasal septum deformation or nasal polyps; respiratory tract infection within 2 weeks prior to screening challenge; pulmonary disease other than mild asthma (defined as controlled by β-2 agonists alone used less than 2 times per week); history of cataracts, glaucoma, or ocular hypertension; receiving escalating doses of allergen immunotherapy within 2 years prior to screening; receiving other medications that might interfere with study measurements or rescue medication; pregnancy; breastfeeding; and participation in another clinical trial within 30 days.

Allergen Challenge and Study Treatments

The ragweed level was maintained at 3500 ± 500 grains/m3 throughout all exposure sessions in the EEU. The pollen concentration was monitored via Rotorod® impact samplers placed in 7 different locations in the room. Samples were taken for 30 seconds at 30-minute intervals, which corresponded with the time points for participant symptom scoring. Participants remained under constant supervision by medical staff inside the unit. A schematic of the EEU is shown in Figure 1.

Participants attended a 3-hour baseline pollen challenge on day 1. Allergen challenge was performed again on days 7 and 14. At the day 1 visit, the site dispensed study medication for the 13 days of treatment along with rescue medication and participants were instructed to record study medication administration time, any missed doses, and any use of rescue medication in a take-home diary. Participants were randomized to either S0597 at doses of 50, 200, or 400 µg/day, or placebo as a nasal spray to be administered once daily in the evening on days 1 to 13. Study medication was taken onsite following the completion of the EEU exposure on day 1 (baseline visit) and day 7. On days 2 to 6 and 8 to 13, participants self-administered the medication once daily at home at approximately 9 PM.

Assessments: Nasal symptoms were monitored at the priming visit, and on baseline day 1, day 7, and day 14, immediately prior to pollen exposure in the EEU and at 6 time points (30, 60, 90, 120, 150, and 180 minutes) during the challenge. Nasal assessments included TNSS, total ocular symptom score (TOSS), total rhinoconjunctivitis symptom score (TRSS), and peak nasal inspiratory flow (PNIF) (See Figure 2). The severity of each component symptom was scored from 0 to 3 and the total scores were calculated by adding the scores for each symptom (Table 1). The average score over the 6 time points during challenge was the test result for each visit.

The primary efficacy end point was TNSS change from baseline to day 14. Secondary efficacy end points were TNSS change from baseline to day 7 and to day 14 in each of the TNSS subscales. Prespecified exploratory efficacy end points were TOSS, TRSS, PNIF, and peak expiratory flow rate (PEFR).

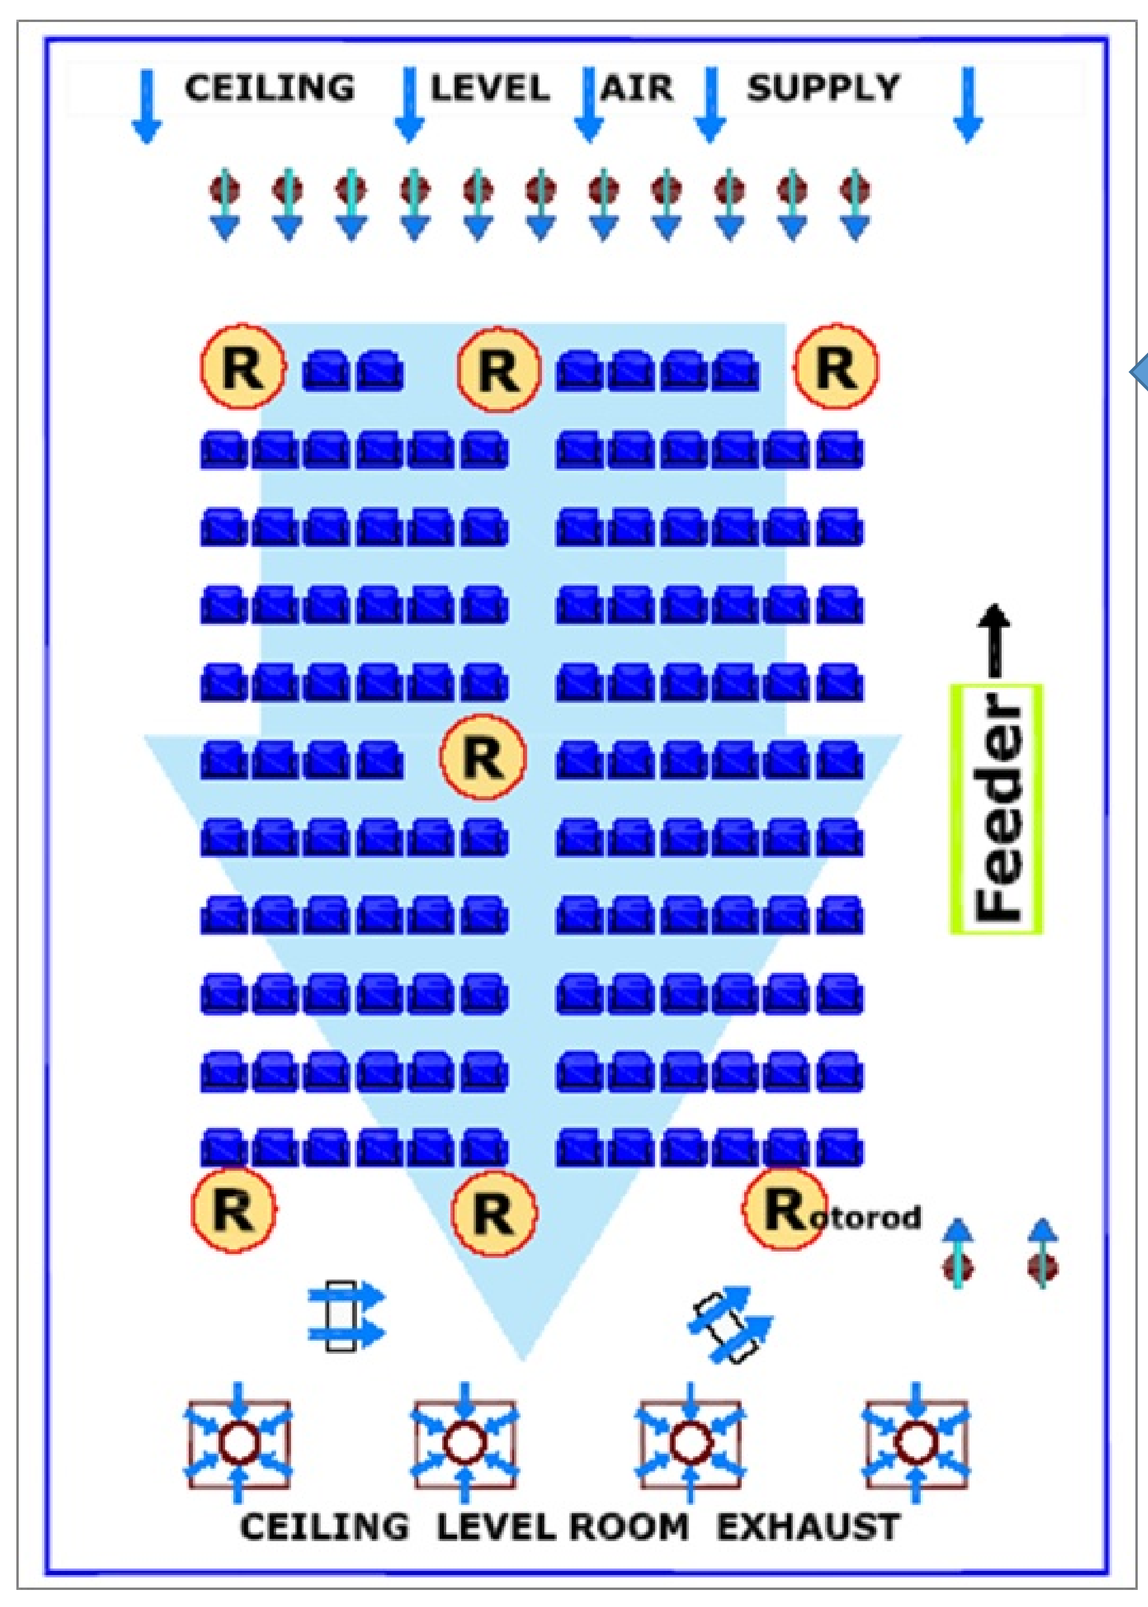
CEILING LEVEL AIR SUPPLY
Feeder
Rotorod
CEILING LEVEL ROOM EXHAUST

Figure 1: Schematic of the EEU
The pollen is dispensed through the feeder on the right and the fans cause a directional flow to allow for the pollen to pass over the seating area and exhaust through the back vents.
® = Rotorod

Figure 2: Symptom Diary Card

The protocol specific diary card is completed by each participant as per the description above every half hour to determine symptom eligibility.

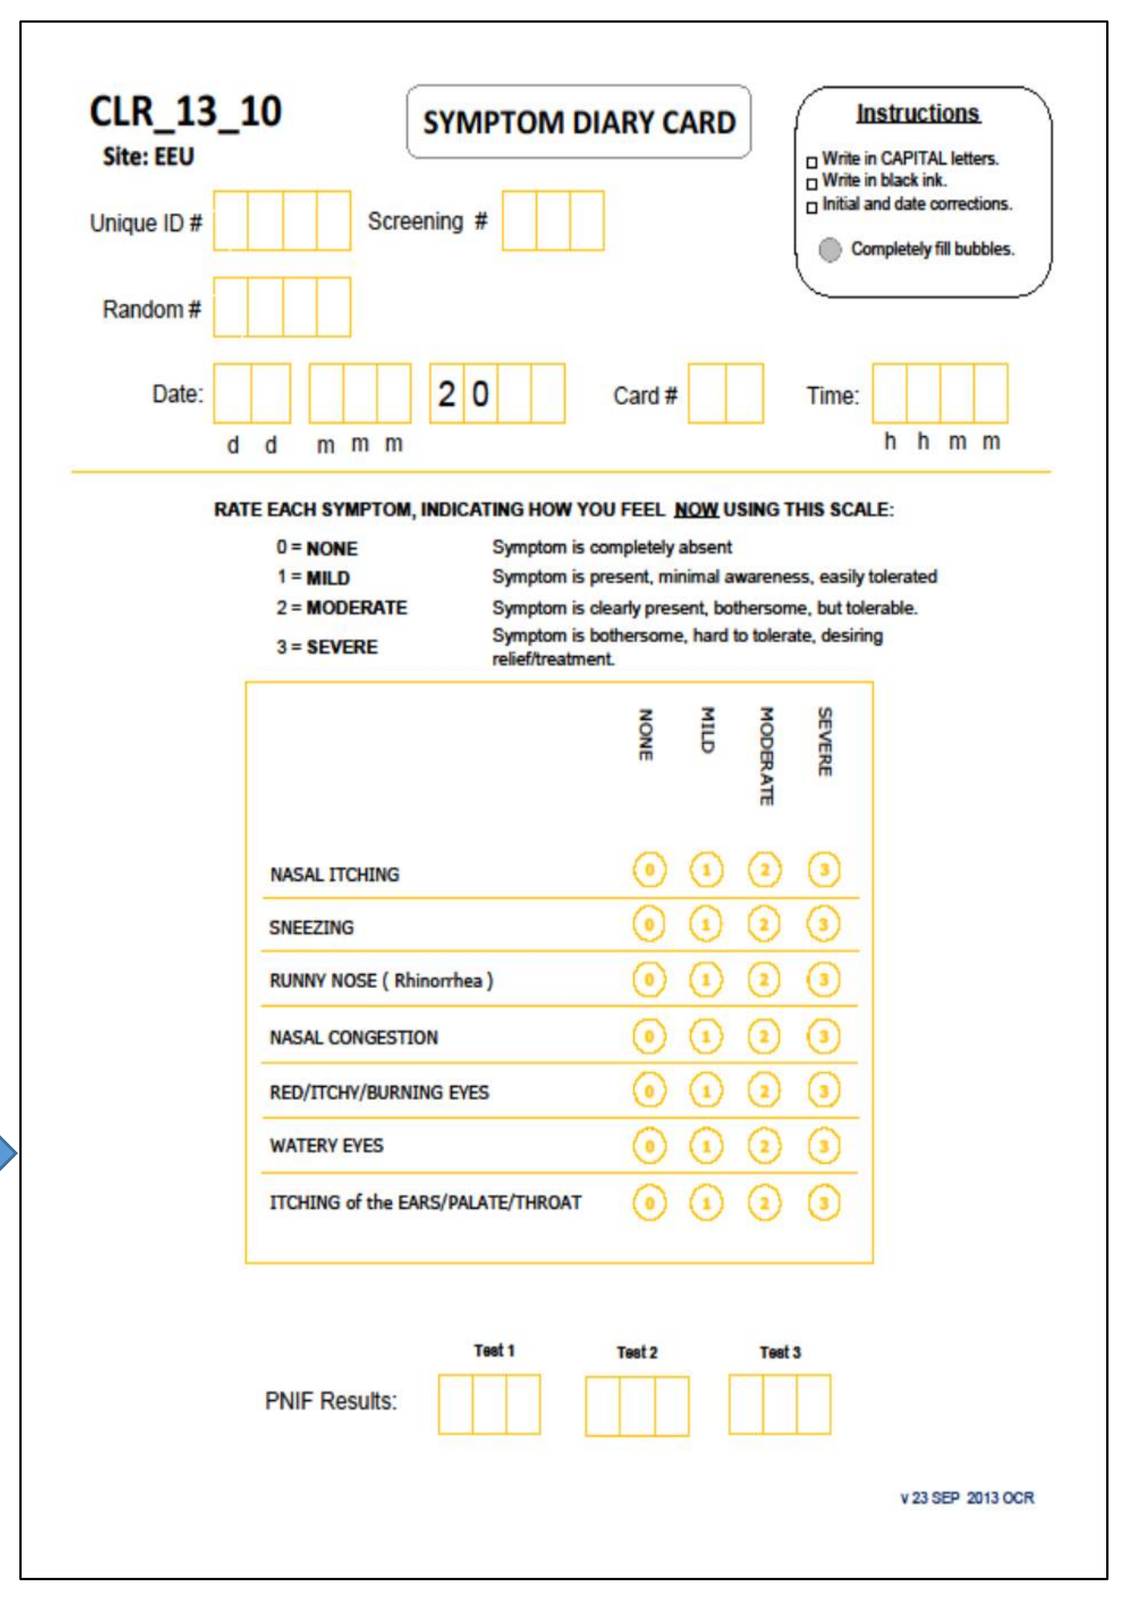
CLR_13_10
Site: EEU

SYMPTOM DIARY CARD

Instructions
Write in CAPITAL letters.
Write in black ink.
Initial and date corrections.
Completely fill bubbles.

Unique ID # Screening #
Random #
Date: d d m m m 2 0 Card # Time: h h m m

RATE EACH SYMPTOM, INDICATING HOW YOU FEEL NOW USING THIS SCALE:
0 = NONE Symptom is completely absent
1 = MILD Symptom is present, minimal awareness, easily tolerated
2 = MODERATE Symptom is clearly present, bothersome, but tolerable.
3 = SEVERE Symptom is bothersome, hard to tolerate, desiring relief/treatment.

	NONE	MILD	MODERATE	SEVERE
NASAL ITCHING	0	1	2	3
SNEEZING	0	1	2	3
RUNNY NOSE (Rhinorrhea)	0	1	2	3
NASAL CONGESTION	0	1	2	3
RED/ITCHY/BURNING EYES	0	1	2	3
WATERY EYES	0	1	2	3
ITCHING of the EARS/PALATE/THROAT	0	1	2	3

PNIF Results: Test 1 Test 2 Test 3

v 23 SEP 2013 OCR

Methods cont'd

Statistical Analyses: Assuming a difference in the change from baseline in TNSS of 1 unit (standard deviation [SD] 1.7) between S0597 and placebo, a sample size of 200 evaluable participants was required to assure at least 80% power at a 2-sided α level of 5%, using a t test statistic for equal group sizes. Approximately 288 participants were to be screened to randomize 220 participants to the study.

The safety population consisted of all randomized participants receiving at least 1 dose of the study treatment. The primary and secondary efficacy analyses were conducted for the intent-to-treat population. Least-squares mean changes in TNSS, TNSS subscales, TOSS, TRSS, PNIF, and PEFR scores from baseline to day 14 were compared among groups using repeated-measures analysis of covariance, with treatment, visit, and treatment-by-visit interaction as fixed effects and baseline value as the covariate (treatment comparison at the 2-sided 0.05 significance level). Pairwise treatment comparisons of each dose of S0597 vs placebo were conducted using the t test. All statistical analyses were performed using SAS 9.3 or subsequent versions (SAS Institute, Cary, North Carolina).

Results

DEOMGRAPHICS		S0597			
	Placebo (n = 56)	50 µg/day (n = 55)	200 µg/day (n = 56)	400 µg/day (n = 55)	Total (n = 222)
Age, years, mean (SD)	39 (10.8)	39 (9.5)	39 (10.9)	38 (10.6)	39 (10.4)
Gender, n (%)					
Female	41 (73.2)	29 (52.7)	36 (64.3)	30 (54.5)	136 (61.3)
Male	15 (26.8)	26 (47.3)	20 (35.7)	25 (45.5)	86 (38.7)
Race, n (%)					
Caucasian	54 (96.4)	51 (92.7)	55 (98.2)	55 (100)	215 (96.8)
Asian	1 (1.8)	1 (1.8)	1 (1.8)	0	3 (1.4)
Other	1 (1.8)	3 (5.5)	0	0	4 (1.8)
Ethnicity, n (%)					
Hispanic	1 (1.8)	0	0	0	1 (0.5)
Non-Hispanic	55 (98.2)	55 (100)	56 (100)	55 (100)	221 (99.5)
Height, cm, mean (SD)[a]	169 (8.0)	170 (9.7)	169 (8.6)	171 (8.2)	170 (8.6)
Weight, kg, mean (SD)[b]	81 (18.2)	87 (22.4)	88 (19.4)	90 (18.9)	87 (19.9)
Body mass index, kg/m², mean (SD)[b]	29 (6.6)	30 (7.7)	30 (7.2)	31 (6.8)	30 (7.1)

Table 1: Demographics of the participants at screening (safety analysis set)

Data are missing for 1 participant in the S0597 50 µg/day group, 1 participant in the placebo group and 1 in the S0597 50 µg/day group. Abbreviation: SD, standard deviation.

A total of 222 participants were randomized into the study and received study medication. Thus, all were included in the population for safety analysis and the intent-to-treat population for the efficacy analysis. The mean (SD) age was 39 (10.4) years, 61.3% were female, and most were Caucasian (96.8%), followed by Asian (1.4%) .

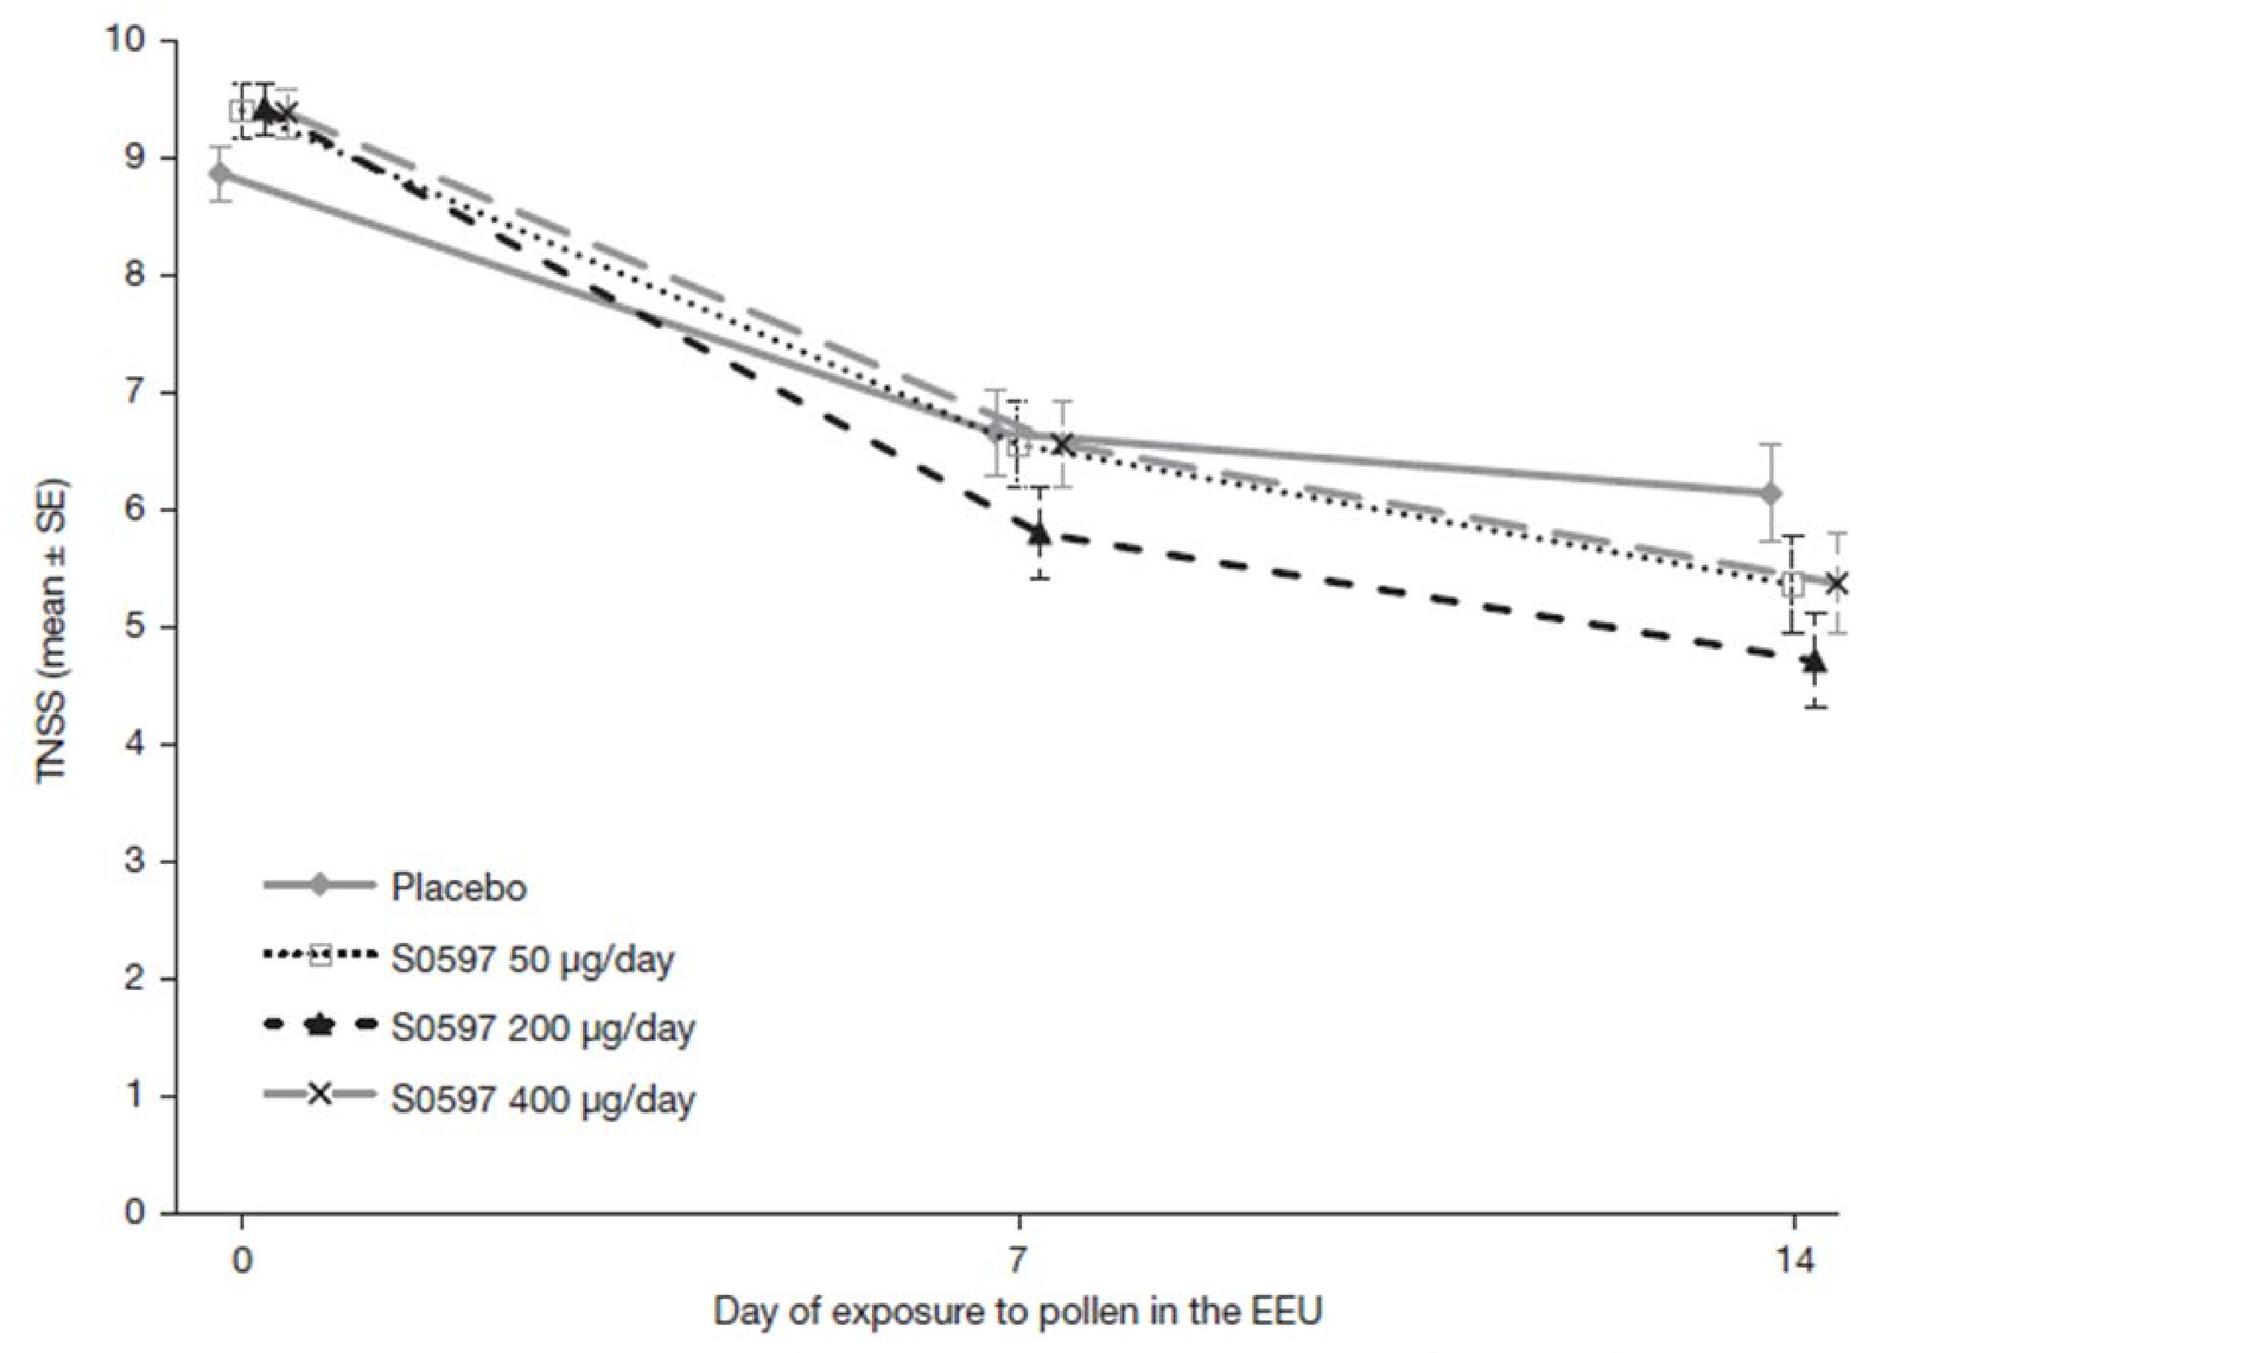
TNSS (mean ± SE)
Placebo
S0597 50 µg/day
S0597 200 µg/day
S0597 400 µg/day
0
7
14
Day of exposure to pollen in the EEU

Figure 3. TNSS over time (intent-to-treat population).
Abbreviations: EEU, environmental exposure unit; SE, standard error; TNSS, total nasal symptom score.
Mean baseline TNSS was between 8.87 and 9.43 across the treatment groups. Analysis of the primary efficacy end point of change in TNSS from baseline to day 14 demonstrated statistically significantly greater improvement in TNSS in all 3 S0597 dose groups compared with placebo. TNSS declined, demonstrating symptom improvement in all treatment groups during the 2-week treatment. The least-squares mean differences in change from baseline between active treatment and placebo were 1.18, 1.84, and 1.17 for the S0597 50, 200, and 400 µg/day groups, respectively; all were statistically significant. None of the exploratory pairwise comparisons between S0597 dose groups reached significance. The greater improvement from baseline in TNSS at day 7 was statistically significant for the S0597 200 µg/day group; least-squares mean difference from placebo 1.25 (95% CI, 0.32–2.18; P = .0088). Similar results were seen for the per-protocol analysis set.

Discussion

This randomized, double-blind, parallel-group, single-center study assessed the dose-related efficacy and safety of 3 doses of S0597, a novel INS, in participants with SAR who were challenged with ragweed pollen in an EEU.

All participants reported nasal symptoms of SAR after pollen challenge and all 3 doses of S0597 (50, 200, 400 µg/day) showed a statistically significant reduction in nasal symptoms (TNSS) from baseline compared with placebo on day 14. The mean improvement in each of the constituent items of the TNSS (rhinorrhea, nasal congestion, sneezing, nasal itching) from baseline to day 14 was also greater in each of the S0597 groups as compared with placebo. There was no evidence that the efficacy response consistently increased with increased dose. The S0597 200 µg/day group demonstrated the most consistent response, with statistically significant improvements in all of the nasal symptoms compared with placebo on days 7 and 14. The difference from placebo for change in the rhinorrhea score was statistically significant with all 3 doses of S0597 on day 14. In addition to these symptom scores, there was a statistically significant improvement in measurements of PNIF (data not shown).

As with other new INS therapies, S0597 was well tolerated at the doses tested. The most common AE in each of the treatment groups, including placebo, was a decrease in blood cortisol levels; the incidence appeared to be somewhat higher among S0597-treated participants compared with those who received placebo, but the event rates were low (2 participants in the placebo group compared with 3 or 4 in each of the S0597 groups). No dose–response relationship was observed among the treatment groups for the incidence of AEs considered severe or for those considered possibly related to study drug.

Conclusion

In conclusion, this study demonstrated that all 3 tested doses of S0597 were effective in relief of nasal symptoms of SAR compared with placebo, and S0597 is a novel intranasal corticosteroid that warrants further development.

Acknowledgements

This study was funded by SunPharma Advanced Research Corporation (Mumbai, India). The authors acknowledge Mr. Daniel Adams and Mr. Barnaby Hobsbawn for their contributions to the successful completion of this study.

UNMATCHED EXPERTISE FROM THE LEADERS IN ENVIRONMENTAL EXPOSURE UNIT TECHNOLOGY SINCE 1985.

Affiliated with Kingston General Hospital and Queen's University.